AAPS 2011

SPX-10624258 and D-Tagatose in the Treatment of Dyslipidemia

Dietrich B. Conze[1], A. Wallace Hayes[1], Claire L. Kruger[1], and Robert A. Lodder[1,2]

1. Biospherics, Incorporated, 6430 Rockledge Drive, Bethesda, MD 20817

2. College of Pharmacy, University of Kentucky, Lexington, KY 40536

Introduction

Dyslipidemia is one of the primary contributors to the development of atherosclerosis. Dyslipidemia is associated with a variety of diseased states including metabolic syndrome, diabetes, obesity, and familial hypercholesterolemia, and is characterized by high levels of triglycerides, increased apolipoprotein (apo) B, high levels of low-density lipoprotein (LDL) cholesterol, and low levels of high-density lipoprotein (HDL) cholesterol. Although the molecular and biochemical mechanisms that contribute to development of dyslipidemia in these diseased states are not completely understood, it is widely accepted that the consumption of diets high in fat, cholesterol, and carbohydrates, especially those enriched with fructose, contribute greatly to the development of disease.

Dietary carbohydrates are absorbed in the small intestine, enter the blood stream, and are absorbed by cells. In negative energy states, carbohydrates are then converted to adenosine triphosphate by glycolysis and the tricarboxylic acid (TCA) cycle, whereas in positive energy states, the excess glycolytic intermediate glycerol 3-phosphate and TCA intermediate citrate accumulate and are converted to triglycerides, packaged into very low-density lipoproteins (VLDLs), and shuttled into the blood stream. Unlike glucose, fructose is primarily metabolized by the liver, but lacks the necessary feedback mechanisms to inhibit subsequent glucose uptake. Thus, the end result in consuming diets high in fructose is a buildup of glycolytic intermediates and the increased production of VLDLs. As the triglycerides in VLDLs are hydrolyzed, the densities of VLDLs decrease, and VLDLs become low-density lipoprotein (LDLs). High-density lipoproteins (HDLs) are synthesized de novo in the blood from the association of apo A-1 and cell-derived phospholipids.

D-tagatose is a C4-epimer of fructose and is converted less efficiently to glycerol 3-phosphate than fructose. Importantly, Biospherics has recently shown that tagatose competitively inhibits fructose absorption in the gastrointestinal tract and, in a phase 2 clinical study, significantly reduced serum triglycerides in type 2 diabetics.

Peroxisome proliferator-activated receptor (PPAR) agonists are well known therapeutics for dyslipidemia. PPARs are nuclear receptors that bind DNA when activated and regulate the expression of genes involved in lipid catabolism and the uptake of oxidized LDLs. SPX-106 is a naturally synthesized PPAR agonist that inhibits the development of dyslipidemia.

The goal of the current study was to determine the effects of D-tagatose and SPX-106 on dyslipidemia and atherosclerosis using low-density lipoprotein receptor (LDLr)-deficient (LDLR-/-) and apolipoprotein E-deficient (apo E-/-) mice.

1.LDLR-/- mice were fed standard chow and gavaged with uncorrelated doses (g/kg/dose) of glucose and fructose (Glu/Fruc) in the presence or absence of tagatose (Glu/Fruc/Tagatose) or a combination of tagatose and SPX-106 (Glu/Fruc/SPX-106T) twice a day for nine weeks.

1.Apo E-/- mice were fed a western diet containing sucrose (341.5 g/kg/diet; TD.88137, Harlan Laboratories) or SPX-106T (341.5 g/kg tagatose + 1 g/kg SPX-106/diet).

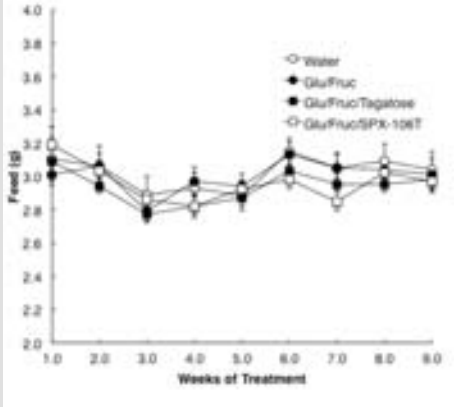

Water
Glu/Fruc
Glu/Fruc/Tagatose
Glu/Fruc/SPX-106T
Feed (g)
Weeks of Treatment

Figure 1. Tagatose and SPX-106T does not affect feed intake in LDLR-/- mice. Values represent the average weekly consumption of standard chow over the course of the treatment period. Error bars represent the standard deviation of the mean.

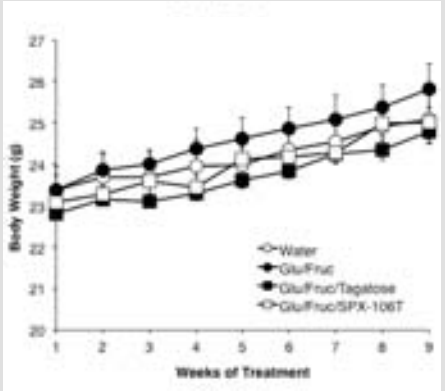

Water
Glu/Fruc
Glu/Fruc/Tagatose
Glu/Fruc/SPX-106T
Body Weight (g)
Weeks of Treatment

Figure 2. Tagatose and SPX-106T does not affect body weight gain in LDLR-/- mice. Mice were feed normal chow and gavaged with uncorrelated doses (g/kg/dose) of glucose and fructose (Glu/Fruc) in the presence or absence of tagatose (Glu/Fruc/Tagatose) or a combination of tagatose and SPX-106 (Glu/Fruc/SPX-106T) for nine weeks. Body weights were recorded weekly and error bars represent the standard deviation of the mean.

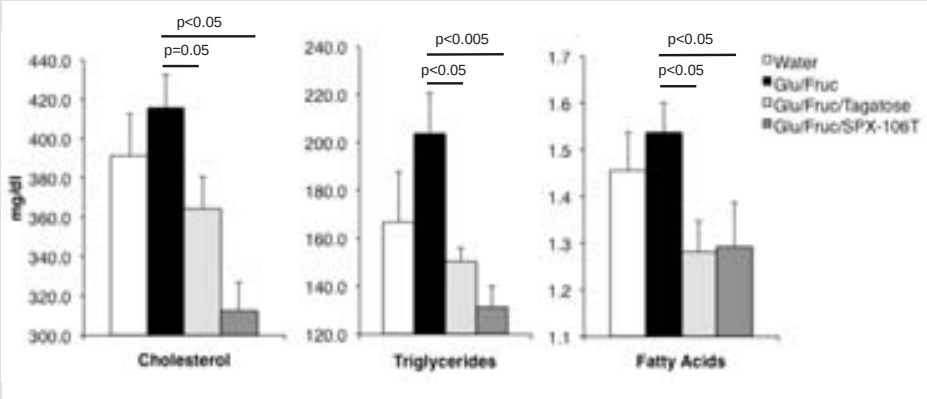

p<0.05
p=0.05
p<0.005
p<0.05
p<0.05
p<0.05
mg/dl
Water
Glu/Fruc
Glu/Fruc/Tagatose
Glu/Fruc/SPX-106T
Cholesterol
Triglycerides
Fatty Acids

Figure 3. Tagatose and SPX-106T reduce serum lipids in LDLR-/- mice. Serum was harvested three weeks after the treatments began and cholesterol, triglycerides, and fatty acids were determined by colorimetric assays. Error bars represent the standard deviation of the mean. P-values comparing the effects between the different groups are noted on the graph.

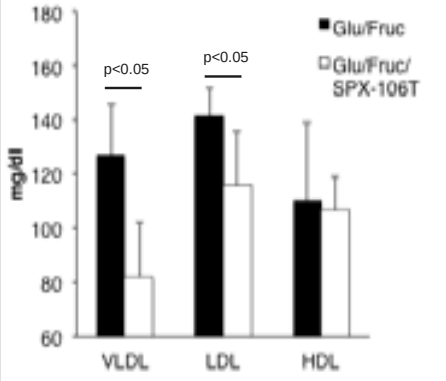

p<0.05
p<0.05
Glu/Fruc
Glu/Fruc/
SPX-106T
mg/dl
VLDL
LDL
HDL

Figure 4. SPX-106T reduces VLDL and LDL in LDLR-/- mice. The amount of VLDLs, LDLs, and HDLs were determined by collecting serum nine weeks after the treatments began, resolving the lipoprotein complexes by FLPC, and quantifying the amount of cholesterol in each FPLC fraction using an enzymatic cholesterol assay. P-values of comparisons of cholesterol between Glu/Fruc/SPX-106T and Glu/Fruc are noted on the graph. Error bars represent the standard deviation of the mean.

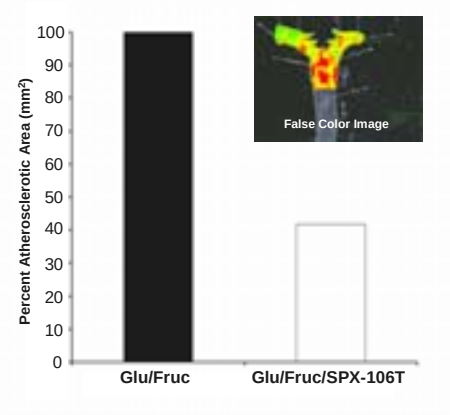

Percent Atherosclerotic Area (mm²)
False Color Image
Glu/Fruc
Glu/Fruc/SPX-106T

Figure 5. SPX-106T reduces atherosclerotic lesions in LDLR-/- mice. Aortic arches were harvested nine weeks after treatments began and the amount of atherosclerotic lesions was determined by false color imaging (inset; red areas indicate locations of the lipid deposits). Lesion area is measured as a fraction of the aortic arch. The percent of atherosclerotic lesions in the SPX-106T treated group was determined by dividing the mean atherosclerotic lesion in the Glu/Fruc/SPX-106T group by the mean atherosclerotic lesion in the Glu/Fruc group.

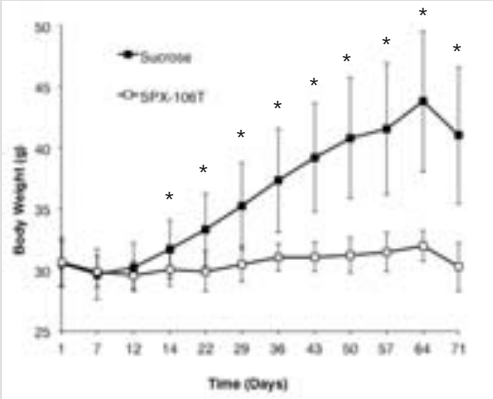

Sucrose
SPX-106T
Body Weight (g)
Time (Days)

Figure 6. SPX-106T prevents body weight gain in apo E-/- mice. Apo E-/- mice were fed standard chow without or with ascending doses of tagatose (increasing 7%/day until 341.5 g/kg tagatose/diet was reached) for 14 days followed by western diet containing sucrose (341.5 g/kg diet) or SPX-106T (341.5 g/kg tagatose + 1 g/kg SPX-106/diet) for eight weeks, respectively. Body weights were measured weekly. Asterisks denote significant differences (p<0.05). Animals were fasted overnight before the day 71 time point.

Group	Kcal/g Diet	Total Feed Consumption (g)	Days of study	Number of Mice	Caloric Intake (Kcal/Mouse/Day)
Sucrose	4.5	3985	70	13	19.71
SPX-106T	3.9	4591	70	13	19.68

Table 1. SPX-106T does not affect feed intake in apo E-/- mice. Apo E-/- mice were fed standard chow without or with ascending doses of tagatose for 14 days followed by western diet containing sucrose or SPX-106T for eight weeks. Daily caloric intake was calculated.

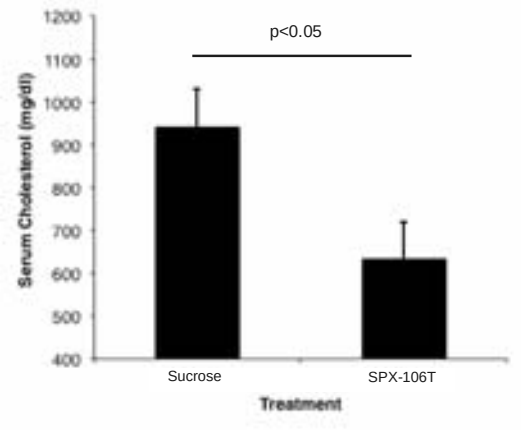

p<0.05
Serum Cholesterol (mg/dl)
Sucrose
SPX-106T
Treatment

Figure 7. SPX-106T prevents hypercholesterolemia in apo E-/- mice. Serum was harvested from the apo E-/- mice fed standard chow without or with ascending tagatose for 14 days followed by a western diet containing sucrose or SPX-106T for eight weeks and total cholesterol was determined by an enzymatic assay.

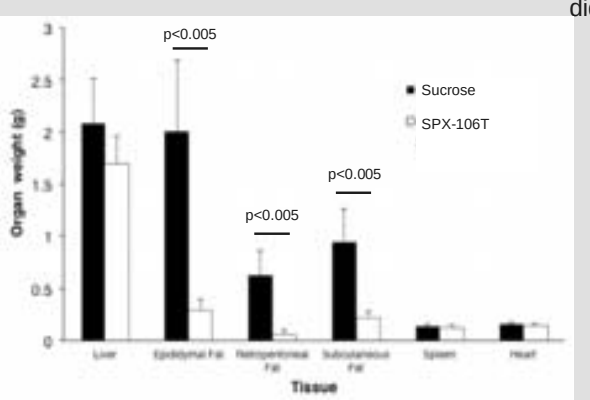

p<0.005
p<0.005
p<0.005
Sucrose
SPX-106T
Organ weight (g)
Tissue

Figure 8. SPX-106T produced reductions in fat deposition in apo E-/- mice. Organs and fatty deposits were harvested from apo E-/- mice fed standard chow without or with ascending tagatose for 14 days followed by a western diet containing sucrose or SPX-106T for eight weeks and weighed.

Results

In LDLR-/- mice

•Tagatose and SPX-106 had no effect on feed intake and weight gain.

•Tagatose and SPX-106T significantly reduced serum cholesterol, triglycerides, and free fatty acids. Karhunen–Loève transformation and multiple linear regression revealed that the effect of SPX-106 was greater than tagatose.

•SPX-106T significantly reduced VLDL and LDL cholesterol by 35 and 17%, respectively.

•SPX-106T reduced atherosclerosis by 57%.

In apo E-/- mice

•SPX-106T prevented weight gain.

•SPX-106T reduced serum cholesterol by 30%.

•SPX-106T significantly reduced the size of fatty deposits and did not affect the size of other organs measured.

Conclusions

•These results are consistent with tagatose inhibiting the conversion of carbohydrates to lipids and SPX-106 promoting lipid catabolism.

•These animal studies suggest that SPX-106T is a promising therapy for the treatment of dyslipidemia.